COMPARATIVE ABSORPTION OF A STANDARDIZED CURCUMINOID MIXTURE AND ITS LECITHIN FORMULATION

John Cuomo[1], Giovanni Appendino[2], Adam S. Dern[1], Erik Schneider[1], Jenna A. Templeton[1], Toni P. McKinnon[1], Mark J. Brown[1], Stefano Togni[3], and Brian M. Dixon[1]

[1]USANA Health Sciences, Inc., 3838 West, Parkway Boulevard, Salt Lake City, Utah 84120, United States

[2]Dipartimento di Scienze Chimiche, Alimentari, Farmaceutiche e Farmacacologiche, Università degli Studi del Piemonte Orientale, Via Bovio 6, 28100, Novara, Italia

[3]Indena S.p.A., Viale Ortles 12, 20139 Milano, Italia

INTRODUCTION

There are three major polyphenols, called curcuminoids, extracted from the turmeric root. More specifically, these are curcumin (1a), demethoxycurcumin (1b), and bisdemethoxycurcumin (1c) (Figure 1). Curcuminoids have been widely used in cooking and in medicine for their antioxidant, anti-inflammatory, and anticancer properties. For example, curcumin inhibits COX2, involved in pain and inflammation; and TNF[1], involved in systemic inflammation associated with several autoimmune disorders. This is only a snapshot of the many endpoints that curcumin targets. Though both *in vitro* and *in vivo* studies have shown great inhibitory activity, even mega-doses of curcumin fail to reach ideal circulating curcumin levels; therefore, recent research efforts have focused on improving the bioavailability of curcumin.

Poor bioavailability stems from the insolubility of curcuminoids in water and lipophilic solvents. Attempts to circumvent solubility issues have been successful in the form of phytosomal formulations. Meriva®* is a 1:2 phytosomal formulation of curcuminoids and lecithin, of which phosphatidylcholine (PC) is a major component. More importantly, PC is amphipathic; it has a positively charged headgroup and two long, neutral tailgroups, allowing for miscibility in both water and lipophilic solvents[2]. Analytical studies suggest that PC completely envelopes a single curcuminoid, so that the hydro- and lipophobic properties of the curcuminoid are eclipsed by the amphipathic nature of PC. Curcuminoids alone are not well absorbed; however, phytosomal complexation with PC allows for easier transportation through phospholipid membranes of cells, thereby greatly improving absorption.

In this study, we compared the bioavailability of three different treatments: two different dosages of Meriva®, and an uncomplexed mixture of the three curcuminoids.

MATERIALS AND METHODS

This study was conducted as a randomized, double-blind, crossover investigation. Nine healthy subjects between the ages of 18 and 55 completed the study. Exclusion criteria included pregnancy, use of prescription medications, gastrointestinal conditions, diabetics, alcohol and/or substance abuse history, and/or allergies to turmeric or curcumin.

Prior to the first study visit, subjects completed an overnight fast and reported to USANA Health Sciences the following morning. Baseline blood samples were drawn before administering one of three treatments: 209 mg Meriva®, 376 mg Meriva®, or 1799 mg uncomplexed curcuminoids. Treatments were taken with a plain bagel with plain cream cheese. Additional blood draws were taken at 2, 4, 8, and 24 h to analyze for plasma curcuminoid concentrations. Following the 4-hour blood draw, an identical meal was given for lunch. Immediately after collection, blood samples were centrifuged and plasma stored at -80˚C until analysis.

After a 7-day washout period, this process was repeated for the two remaining study visits. Subjects were administered a different treatment than the previous visit, until all three treatments were administered.

STRUCTURES OF THE THREE MAIN CURCUMINOIDS

	R_1	R_2
1a Curcumin	OMe	OMe
1b Demethoxycurcumin	OMe	H
1c Bisdemethoxycurcumin	H	H

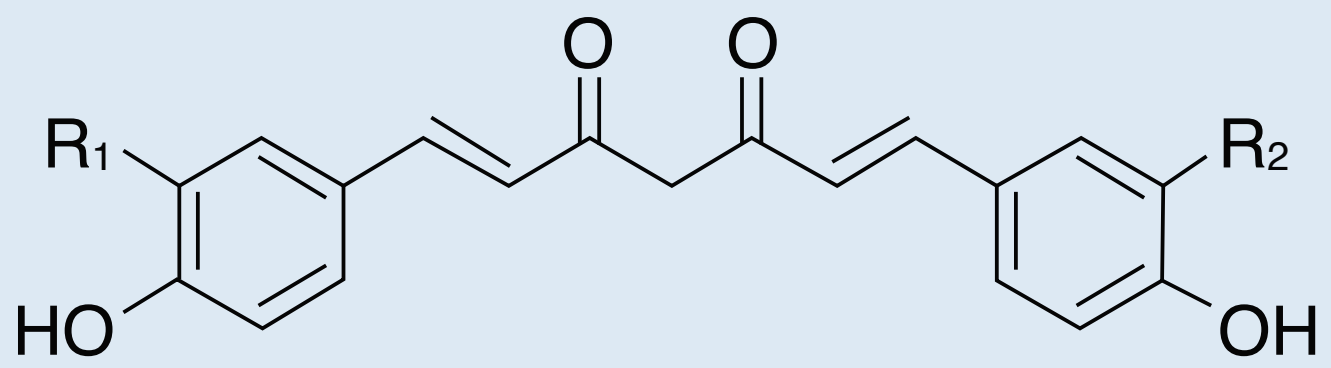

Figure 1. Structure of the three main curcuminoids. The three main curcuminoids found in turmeric differ at two points on each ring. Substituting the corresponding groups for R1 and R2 yields the appropriate structure for each curcuminoid.

Determination of Curcuminoid Concentration by HPLC-MS/MS

The HPLC-MS/MS procedure used was adapted from Liu et al[3]. Stock solutions of each curcuminoid were prepared and these were further diluted with methanol to yield standard solutions of 0.5, 2.5, and 50 ng/50 µL. In order to compensate for matrix effects, a calibration curve was made for each subject by spiking baseline plasma with the appropriate standard to achieve concentrations of 5, 250, and 500 ng/mL. These calibration standards were run multiple times between samples, resulting in several calibration curves that also served as a system suitability check. The analysis was carried out using 2-propanol/0.03% formic acid (35:65, v/v) as mobile phase, with an injection volume of 10 µL, a run time of 15 min, and a flow rate of 1.0 mL/min. The transitions monitored were m/z 369.2 to 285.2 for curcumin (1a), 339.2 to 255.0 for demethoxycurcumin (1b), and 309.1 to 225.0 for bisdemethoxycurcumin (1c) (Figure 1).

Sample Preparation

A 0.2-mL aliquot of plasma was transferred to a clean microcentrifuge tube and treated with 100 µL of a solution containing 1000 U of β-glucuronidase in 0.1 M phosphate buffer (pH 6.86) and 40 µL of methanol. The resulting mixture was then thoroughly vortexed and incubated at 37°C for 1 h to hydrolyze phase-2 conjugates of curcuminoids. After incubation, curcuminoids were extracted with 1 mL of ethyl acetate, and the mixture was vortexed for 1 min, followed by sonication in a water bath for 15 min. After centrifugation at 15,000 x g for 6 min, the upper organic layer was transferred to a microcentrifuge tube and evaporated to dryness at 30°C under negative pressure in a centrifugal concentrator. This extraction process was repeated twice. The dried extract was reconstituted in 100 µL methanol and 10 µL was injected into the HPLC-MS/MS.

RESULTS

- **Curcumin was 18 times more bioavailable in the Meriva® formulation than the reference (Figure 2A).**
- **Meriva® yielded demethoxycurcumin and bisdemethoxycurcumin plasma concentrations 50- to 60-fold higher than that of the reference (Figures 2B and 2C).**
- **Overall curcuminoid absorption was 29 times higher for Meriva® than the reference (Figure 2D).**
- **Meriva® was absorbed twice as fast as the reference (Table 1).**

DISCUSSION & CONCLUSION

The purpose of this study was to prove whether or not complexation with a phospholipid improves human absorption and the pharmacokinetic profile of curcuminoids. A randomized, double-blind, crossover study was carried out with nine

TABLE 1. AREA UNDER THE CURVE (AUC), c_{MAX}, t_{MAX}, AND RELATIVE ABSORPTION FOR EACH TREATMENT OF CURCUMINOIDS[a].

Curcuminoid	Formulation	AUC (ng/mL)	c_{max} (ng/mL)	t_{max} (h)	Relative absorption[b]
Curcumin	Meriva® high Meriva® low Reference	538.0 ± 130.7 272.6 ± 68.52 122.5 ± 29.3	50.3 ± 12.7 24.2 ± 5.9 9.0 ± 2.8	3.8 ± 0.6 4.2 ± 0.8 6.9 ± 2.2	19.2[c] 17.5[c] 1.00
Demethoxycurcumin	Meriva® high Meriva® low Reference	655.0 ± 195.7 297.4 ± 107.3 55.8 ± 15.5	134.6 ± 40.6 39.1 ± 11.4 4.2 ± 1.1	2.4 ± 0.3 3.1 ± 0.4 4.4 ± 1.0	68.3[d] 55.5[d] 1.00
Bisdemethoxycurcumin	Meriva® high Meriva® low Reference	142.2 ± 58.2 70.1 ± 34.3 24.6 ± 10.3	24.9 ± 8.1 8.8 ± 3.1 2.1 ± 0.8	2.2 ± 0.4 2.4 ± 0.6 3.4 ± 1.2	56.8[e] 51.3[e] 1.00
Total curcuminoids	Meriva® high Meriva® low Reference	1336.0 ± 357.1 640.2 ± 197.7 202.8 ± 53.8	206.9 ± 54.9 68.9 ± 16.9 14.4 ± 4.2	2.7 ± 0.3 3.3 ± 0.3 6.9 ± 2.2	31.5[f] 27.2[f] 1.00

[a]Actual results not baseline subtracted and errors are SEM; [b]AUC normalized; [c]Average: 18.3; [d]Average: 61.9; [e]Average: 54.1; [f]Average: 29.4.

volunteers, measuring the plasma concentrations of three curcuminoids after supplementation with three different treatments: two dosages of Meriva® and one dosage of the same batch of curcuminoid mixture (used for the formulation with lecithin). The dosages administered were inspired by previous clinical studies for inflammatory conditions, where active dosages of around 1-2 g/day of unformulated curcuminoid mixtures, and around 200-300 mg/day of curcuminoids as Meriva® were used[4].

Free curcumin (1a) could not be detected in any plasma samples. This is in accordance with previous studies that have mostly failed to detect unconjugated curcumin in human plasma, even after the administration of mega-doses of curcumin[5]. The peak plasma total curcuminoid concentration (c_{max}) reached with the high dosage of Meriva® was 206.9 ± 164.7 ng/mL, and the corresponding time of the peak plasma curcuminoid concentration (t_{max}) was reached at 2.7 ± 1 h after the administration.

From these data, the average absorption of curcumin (1a) was calculated to be about 18-fold higher from Meriva® than from the corresponding reference. Moreover, the overall curcuminoid absorption was about 29-fold higher for Meriva® compared to the unformulated reference, since the plasma concentration of demethoxycurcumin (1b) and bisdemethoxycurcumin (1c) from intake of Meriva® were 50- to 60-fold higher than from the corresponding unformulated curcuminoid mixture. Remarkably, the major plasma curcuminoid was demethoxycurcumin (1b) and not curcumin (1a) with both dosages of Meriva® investigated. The marked differences in the plasma curcuminoid profile could not be accounted for by the nature of the starting materials, since Meriva® capsules and the uncom-

MERIVA YIELDS SUPERIOR CURCUMINOID BIOAVAILABITY

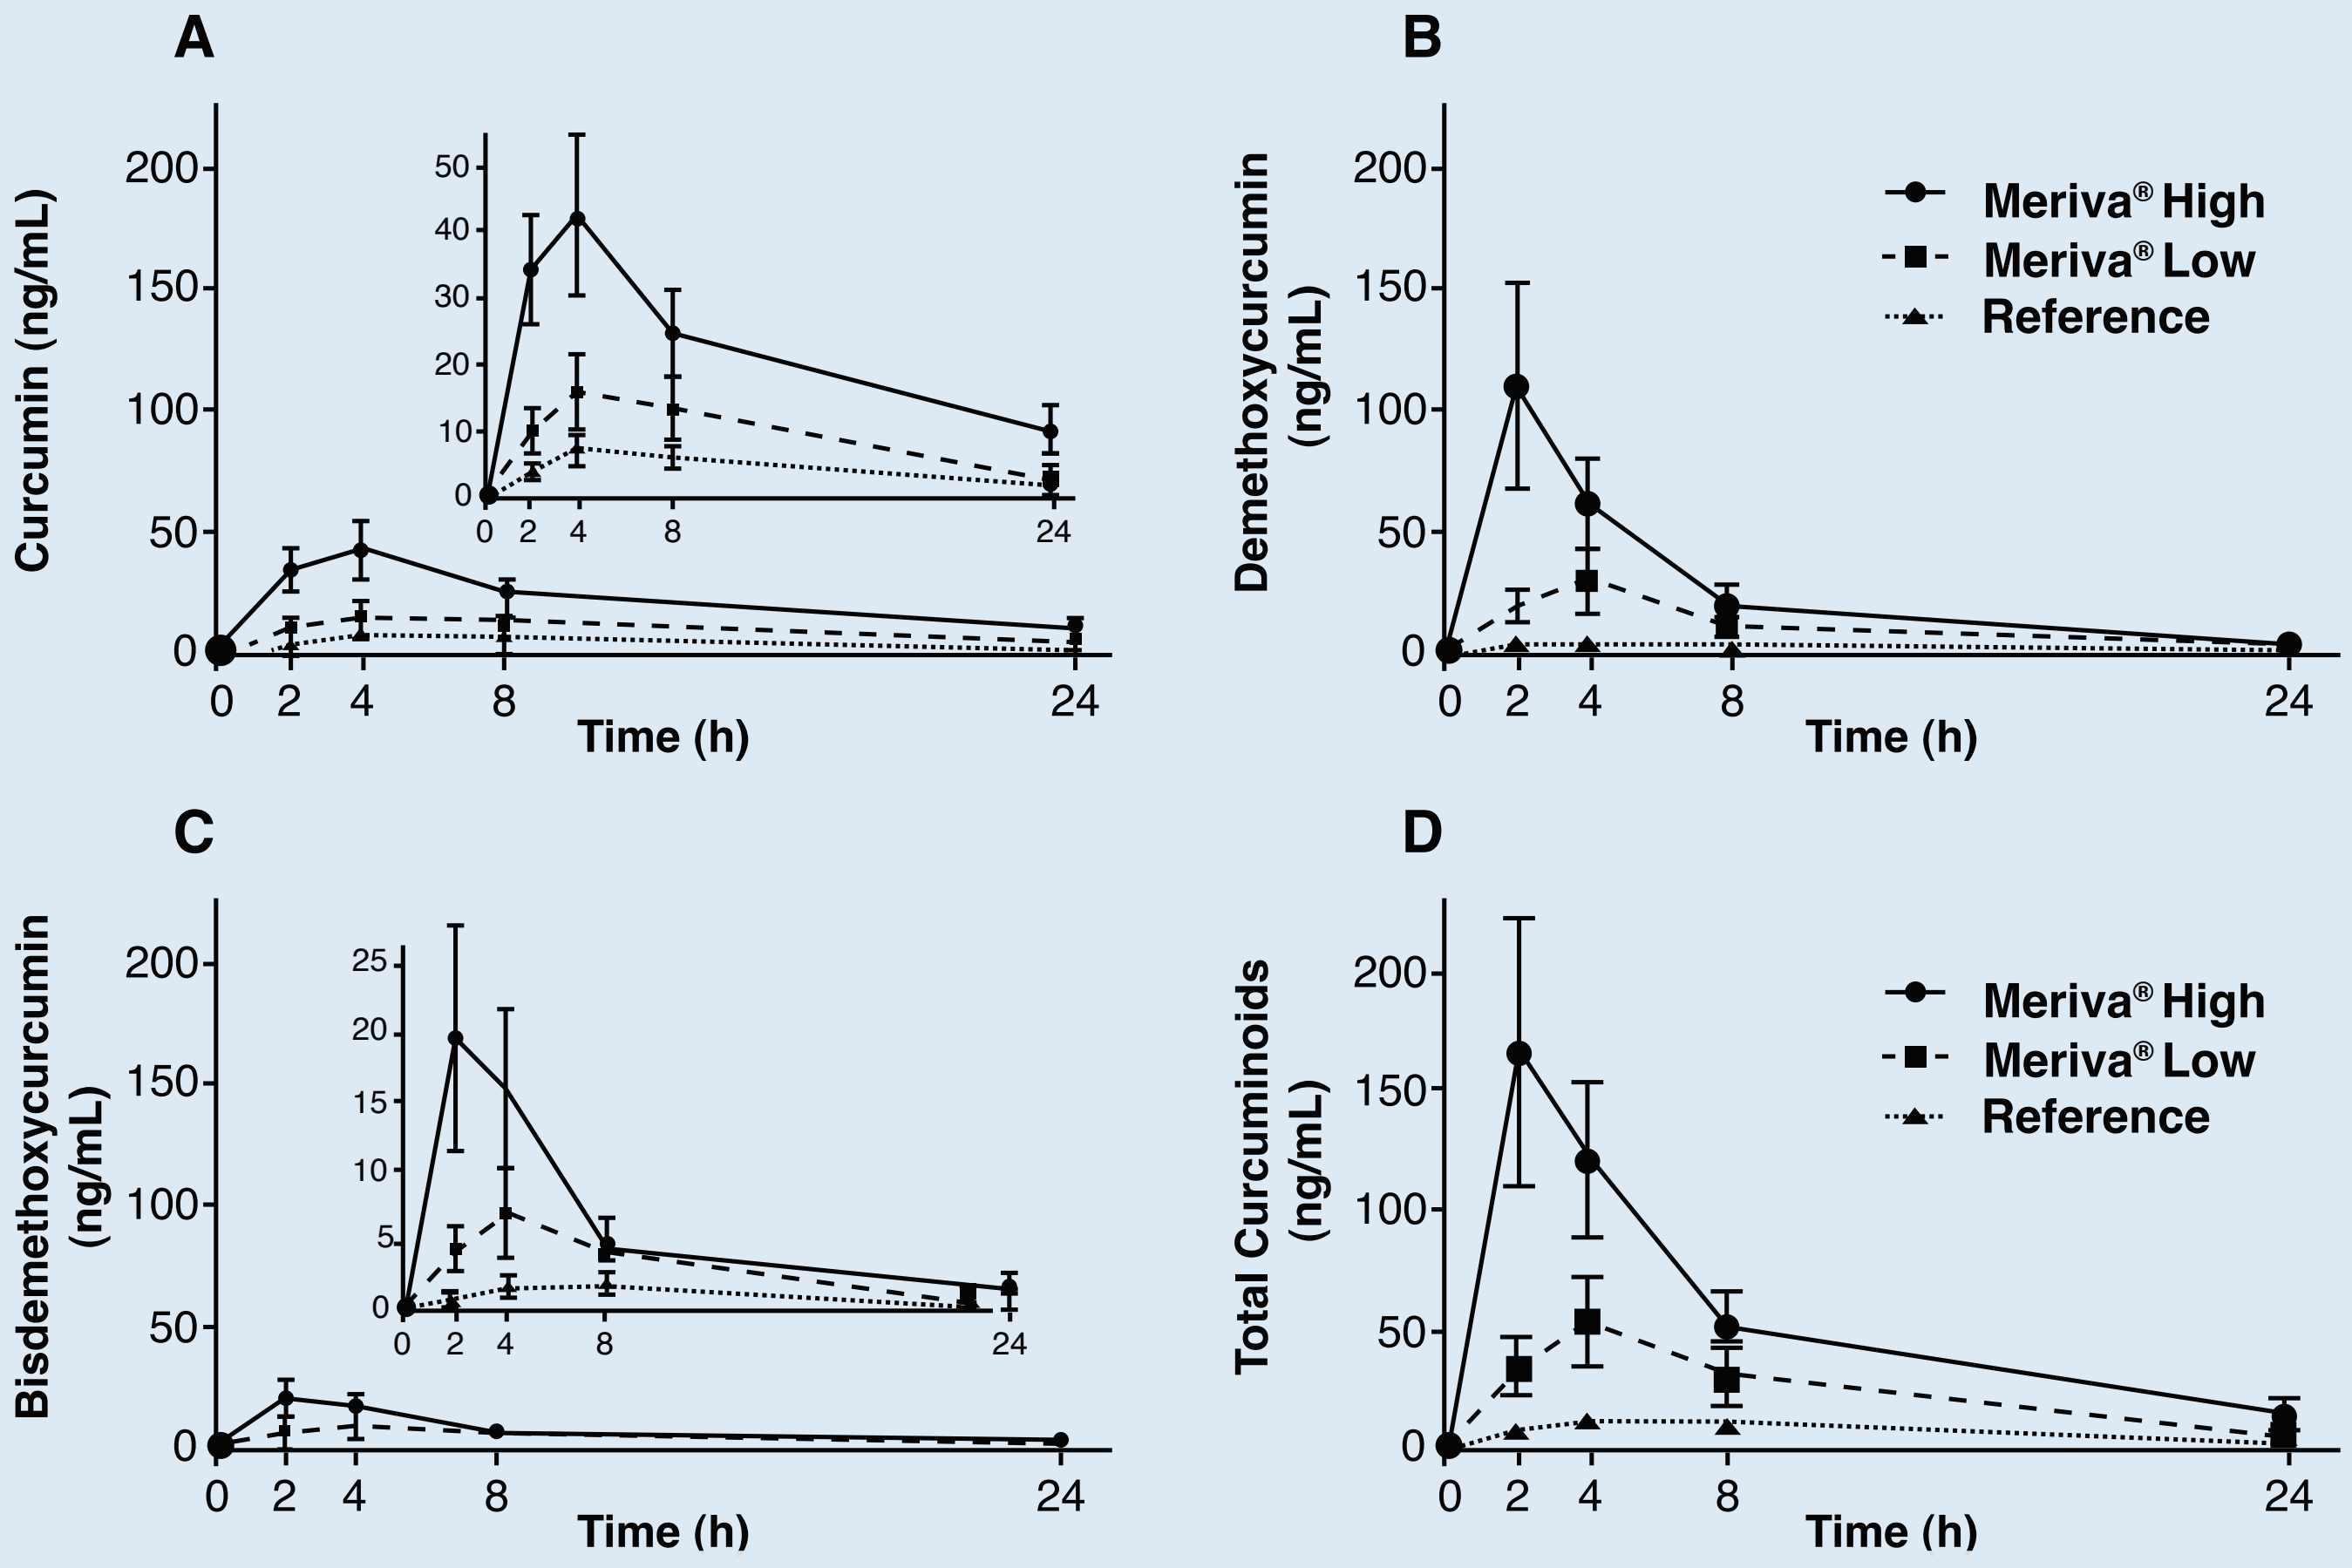

A
Curcumin (ng/mL)
B
Demethoxycurcumin (ng/mL)
C
Bisdemethoxycurcumin (ng/mL)
D
Total Curcuminoids (ng/mL)
Time (h)
Meriva® High
Meriva® Low
Reference

Figure 2. Pharmacokinetic data for curcumin, demethoxycurcumin, bisdemethoxycurcumin, and total curcuminoids for each dosage. Concentrations are expressed in ng/mL and refer to enzymatically hydrolyzed plasma samples. Circles represent high dosages of Meriva, squares represent low-dose Meriva, and triangles represent the reference materical. Inserts (A and C) show an expanded view of the original data. The data shown are baseline subracted means ± SEM.

plexed curcumin capsules contained very similar curcuminoid profiles. It is now obvious that demethoxylated forms of curcumin (1b and 1c) have a better intrinsic absorption than curcumin (1b) and that formulation with phospholipids increases these differences in bioavailability. Interestingly, turmeric is often used in cuisine associated with lecithin-rich ingredients like eggs or vegetable oils, and these observations might also hold well for the dietary intake of curcuminoids.

The reasons for this unexpected increase in the plasma concentrations of the demethoxylated curcuminoids (1b and 1c) over curcumin (1a) might be involved in a process not unlike the one that generates enterolactone and enterodiol from flax lignans[6]. The hydrolytic stabilization of curcumin at intestinal pH might in fact translate into a significant curcumin load for the gut microflora, known to reductively demethoxylate dietary phenolics. The possibility that demethoxycurcumin (1b) and bisdemethoxycurcumin (1c) are generated from curcumin (1a) by liver metabolization seems unlikely, since dietary phenolics are generally *oxidatively O-demethylated* rather than *reductively C-demethoxylated* by liver enzymes. Furthermore, phase-2 metabolization to conjugates is generally the primary metabolic pathway for dietary phenolics[7]. The unique plasma curcuminoid profile might play a role in the clinical efficacy of Meriva® at dosages much lower than those of unformulated curcumin, since demethoxycurcumin is more potent than curcumin in many molecular assays of anti-inflammatory activity[8]. The presence of demethoxylated curcuminoids in most "curcumin" samples has, surprisingly, been largely overlooked, and the fragmentary state of our knowledge on the *in vivo* biological profile of these compounds makes it difficult to evaluate the clinical meaning of differences in the plasma curcuminoid profile. However, based on *in vitro* studies, a better anti-inflammatory curcuminoid profile seems possible for Meriva® compared to unformulated curcuminoid mixtures. This would certainly be worthy of further evaluation. Our data add to the growing body of information suggesting that curcuminoids have different biological profiles. This draws attention to their different bioavailabilities and the need to specify the composition of "curcumin" whenever this compound is used in both cellular and clinical studies.

Curcumin has been a sort of "forbidden fruit" for biomedical research[1], since its poor oral bioavailability has substantially hampered clinical development, despite the very promising indications of the preclinical research[9]. We have demonstrated that formulation with phospholipids[10] improves the human absorption of curcuminiods, without however, leading to pharmacologically active plasma concentrations and with only phase-2 metabolites being detectable[6]. While phase-2 metabolites might play a role *in vivo*, either as prodrugs or as targeting agents[11], the failure to reach pharmacologically active plasma curcuminoid concentrations even with clinically validated dosages of Meriva®[12], raises the issue of how to evaluate effective dosages of multi-targeted agents whose action *in vivo* might be the result of the combinatorial binding to several protein targets and/or the epigenetic modulation of their expression[13].

References

[1] Goel A, et al. Mol Nutr Food Res. 2008; 52: 1010-1030.
[2] Kidd PM. Altern Med Rev. 2009; 14: 226-246.
[3] Liu, et al. J Pharm Biomed Anal. 2006; 40: 720-727.
[4] Keith C, et al. Nat Rev Drug Discovery. 2005; 4: 71-78.
[5] (a) Bisht S, et al. Curr Drug Discovery Tecnol. 2009; 192-199.
(b) Anand P, et al. Mol Pharmaceutics. 2007; 4:807-818.
(c) Lao CD, et al. BMC Complement Altern Med. 2006; 6: 10.
[6] Lampe JW. J Nutr. 2003; 133: 956S-964S.
[7] Crozier A, et al. Nat Prod Rep. 2009; 26: 1001-1043.
[8] (a) Zhang L, et al. Int Immunopharmacol. 2010; 10: 331-338.
(b) Luthra PM, et al. Biochem Biophys Res Commun. 2009; 284: 420-425.
(c) Ahmed T, et al. Pharmacol Biochem Behav. 2009; 91: 554-559.
(d) Yodkeere S, et al. J Nutr Biochem. 2009; 20: 87-95.
(e) Zhang L, et al. Neurosci Lett. 2008; 447: 48-53.
[9] (a) Epstein J, et al. Br J Nutr. 2010; 103: 1545-1557.
[10] Duke JA. CRC Handbook of Medicinal Spices;
CRC Press: Boca Raton, FL, 2003; pp. 137-144.
[11] (a) Roycik MD, et al. Curr Pharm Des. 2009; 15: 1295-1308.
(b) Nakayama SK. Methods Enzymol. 2005; 400: 263-272.
[12] (a) Belcaro G, et al. Panminerva Med. 2010; 52: 55S-62S.
(b) Allegri P, et al. Clin Opthalmol. 2010; 4: 41-46.
(c) Belcaro G, et al. Altern Med Rev. 2010; 15: 337-344.
[13] Fu S, et al. Cancer. 2010; 116: 4670-4676.¬¬

***Meriva® is a tradmark of Indena S.p.A.**

These results have been published:
Cuomo, J., Appendino, G., Dern, A.S., Schneider, E., McKinnon, T.P., Brown, M.J., Togni, S., Dixon, B.M. (2011) J. Nat. Prod. Epub published online: March 17, 2011.

* These statements have not been evaluated by the Food and Drug Administration. This product is not intended to diagnose, treat, cure, or prevent any disease.